Heparosan glyco-conjugates of rFVIII and rFIX show prolonged circulating half-life in haemophilia mouse models

ISTH15ABS-3457

C Behrens[1], J Buchardt[1], M L Broberg[1], D M Karpf[1], M Loftager[1], M S Jensen[1], H W Bagger[1], M Thórólfsson[1], M Dainiak[1], J Karlsson[1], E B Hansen[1], J Krarup[1], H Rahbek-Nielsen[1], M Kjalke[1], P L DeAngelis[2], F M Haller[2], J Haaning[1]

[1]Novo Nordisk A/S, Måløv, Denmark, [2] Caisson Biotech, LLC, Oklahoma City, OK, USA

Objective

- Evaluate the possibility of increasing half-life of rFVIII and rFIX by conjugation to heparosan.

Conclusions

- Heparosan was conjugated to the O-linked glycan in the B-domain of FVIII and an N-linked glycan in the activation peptide of FIX
- Activation resulted in FVIIIa and FIXa similar to unconjugated FVIIIa and FIXa
- The terminal half-life in knock-out mice were prolonged 1.8-fold for N8-HEP and 2.5-fold for N9-HEP
- **The data suggest that heparosan can be considered for half-life prolongation of FVIII and FIX**

Introduction

- Several strategies for increasing the half-life of recombinant FVIII and FIX are currently being pursued to improve prophylaxis in haemophilia. One concept involves polymer conjugation.
- Among several polymers evaluated, heparosan[1] was selected for conjugation to rFVIII and rFIX.
- Heparosan is a linear polysaccharide containing repeating units of glucuronic acid (GlcUA) and N-acetylglucosamine (GlcNAc) (Figure 1).

Figure 1 Structure of heparosan

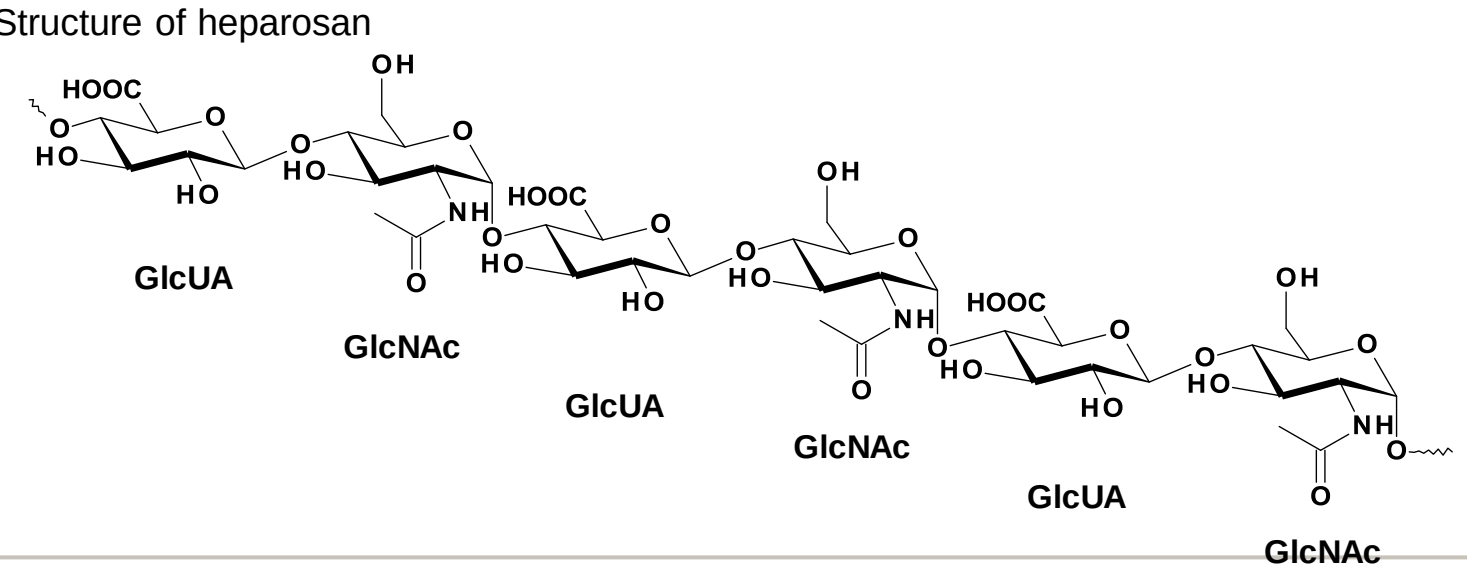
GlcUA
GlcNAc
GlcUA
GlcNAc
GlcUA
GlcNAc

Methods

- 40kDa heparosan was glycoconjugated to the O-linked glycan in the 21 amino acid B-domain of turoctocog alfa (N8-HEP) and to an N-linked glycan in the activation peptide of FIX (N9-HEP) as described[2,3]
- SDS-PAGE with and without activation: N8-HEP (12.5 IU) was cleaved 5 min at 37ºC with 2 U/mL human α-thrombin (Boehringer Mannheim). Samples were incubated 10 min at 70ºC in SDS-PAGE sample buffer with reducing agent before SDS-PAGE using a 7 % TA gels and Tris-Acetate running buffer (NuPage). N9-HEP (2.8 µg) was cleaved 6 h at room temperature with 2.5 nM FXIa (Enzyme Research Lab). SDS-PAGE of reduced samples were analysed on a 4-12 % BIS-TRIS gel using MOPS running buffer(NuPage)
- Pharmacokinetics (PK)
 - FVIII exon 16 knock-out (*F8*-KO) mice were dosed 140 IU/kg N8-HEP or Advate®, and FIX knockout (*F9*-KO) mice were dosed 290 IU/kg N9-HEP or 360 IU/kg BeneFix® intravenously in the tail vein.
 - Blood was sampled from the orbital plexus under Isofluran/O_2/N_2O with three samples per mouse and three to four samples per time point in the time range of predose to 168 h post administration.
 - The citrate-stabilized blood was diluted 5-fold in buffer, centrifuged and the supernatant isolated.
 - Samples were analysed for FVIII or FIX activity in chromogenic assays (Coatest SP, Chromogenix, and Biophen® FIX, Hyphen Biomed, respectively.
 - Pharmacokinetic parameters were estimated by non-compartmental analysis (NCA) using Phoenix Pharsight software. Half-life (t½) was estimated on the terminal part of the curve, clearance (CL) and the mean residence time (MRT) were extrapolated to infinity.

References

1. DeAngelis PL Espert Opin Drug Deliv 2015; 12(3):349-352.
2. Stennicke HR et al. Blood 2013; 121(11): 2108-2116.
3. Østergaard H et al. Blood 2011; 118 (8): 2333-2341.

Presented at the International Society on Thrombosis and Haemostasis (ISTH) Annual Meeting, 20–25 June 2015, Toronto, Canada.

Results

FVIII:

- Heparosan was conjugated to the O-linked glycan in the truncated B-domain of turoctocog alfa (Figure 2).
- SDS-PAGE analysis demonstrated that the presence of the heparosan polymer on the heavy chain. After thrombin cleavage, FVIIIa from N8-HEP was identical to FVIIIa from unconjugated rFVIII (Figure 3)
- PK in *F8*-KO mice demonstrated a 1.8-fold prolongation of half-life compared to rFVIII (Figure 4 and Table 1)

FIX:

- Heparosan was conjugated to an N-linked glycan in the activation peptide of recombinant FIX (Figure 5).
- SDS-PAGE demonstrated the presence of heparosan. After FXIa cleavage, FIXa from N9-HEP was identical to FIXa from unconjugated rFIXa (Figure 6)
- PK in *F9*-KO mice demonstrated a 2.5-fold prolongation of half-life compared to rFIX (Figure 7 and Table 2)

N8-HEP

Figure 2 Structure of N8-HEP before and after thrombin cleavage

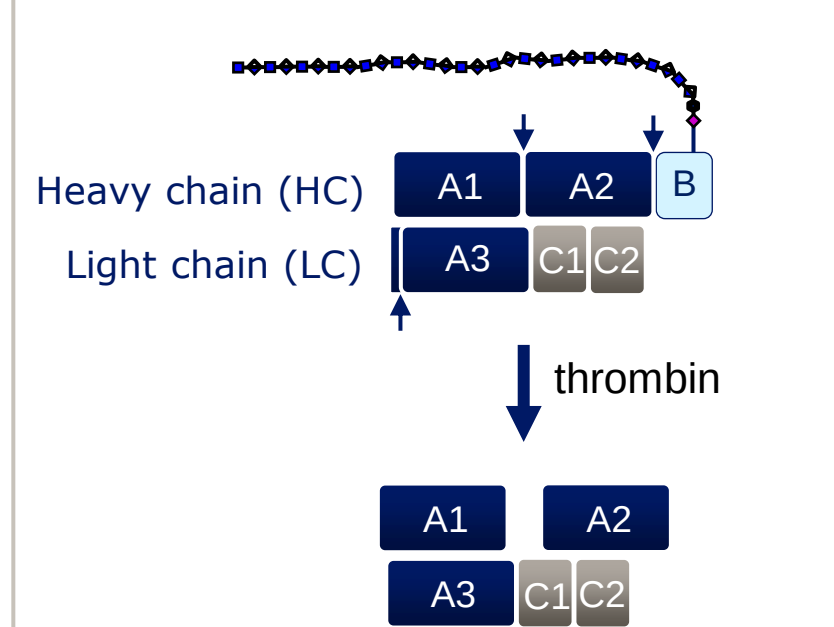
Heavy chain (HC)
Light chain (LC)
A1
A2
B
A3
C1
C2
thrombin
A1
A2
A3
C1
C2

Figure 3 SDS-PAGE of N8-HEP with and without thrombin cleavage

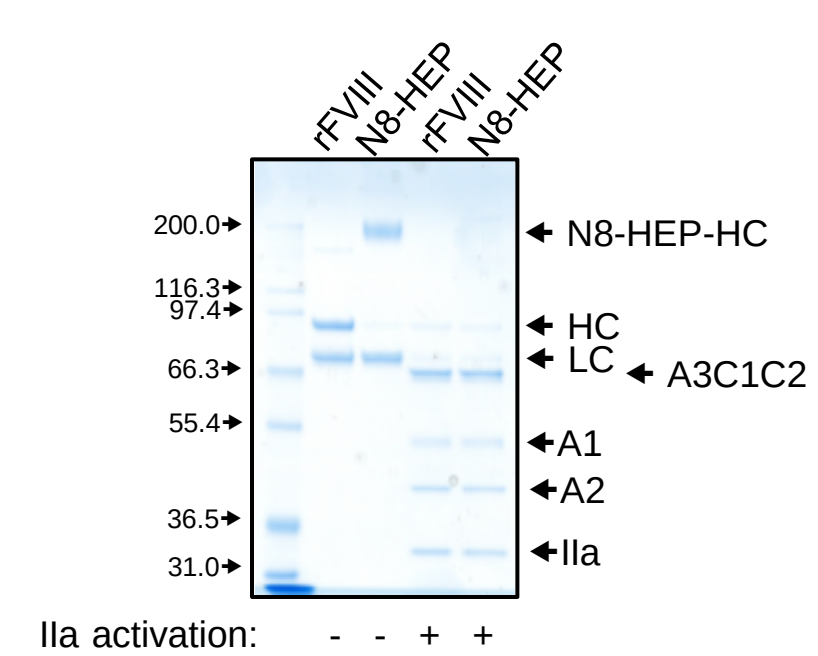
rFVIII
N8-HEP
rFVIII
N8-HEP
200.0
116.3
97.4
66.3
55.4
36.5
31.0
N8-HEP-HC
HC
LC
A3C1C2
A1
A2
IIa
IIa activation: - - + +

Figure 4 FVIII activity in blood samples from *F8*-KO mice dosed i.v. with 140 IU/mL of FVIII.

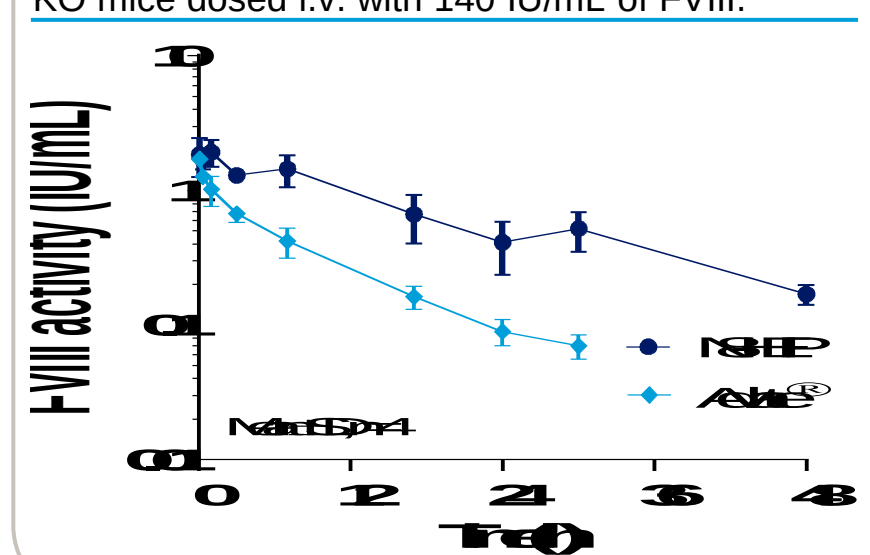
FVIII activity (IU/mL)
0
12
24
36
48

Table 1 Estimated PK parameters

rFVIII	PK parameters		
	t½ (h)	CL (mL/h/kg)	MRT (h)
N8-HEP	14	3.7	20
Advate®	8.1	14	10

N9-HEP

Figure 5 Structure of N9-HEP before and after FXIa cleavage

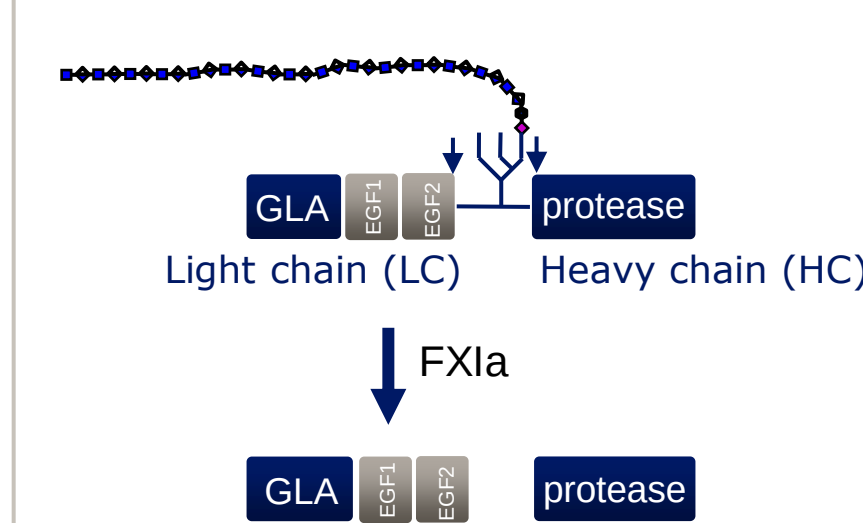
GLA
EGF1
EGF2
protease
Light chain (LC)
Heavy chain (HC)
FXIa
GLA
EGF1
EGF2
protease

Figure 6 SDS-PAGE of N9-HEP with and without FXIa cleavage

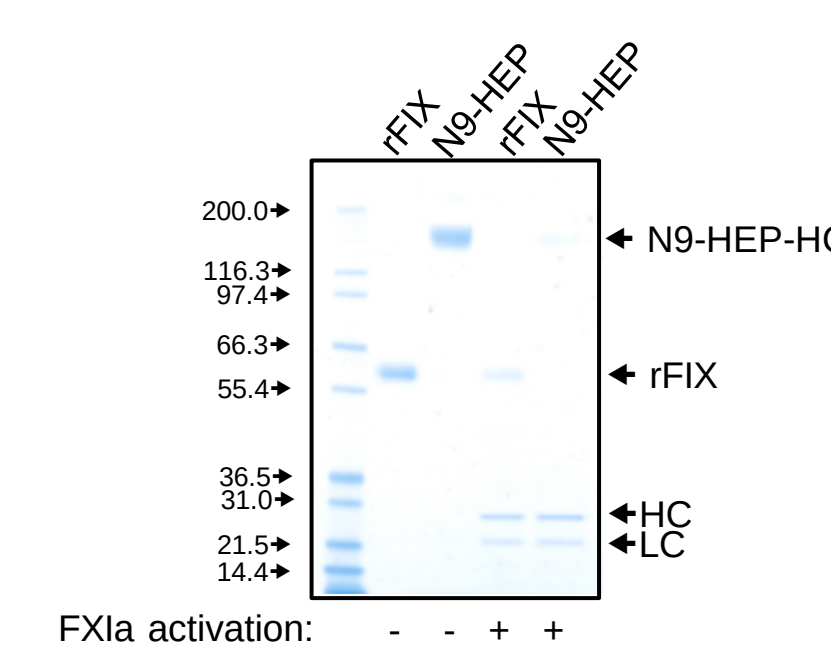
rFIX
N9-HEP
rFIX
N9-HEP
200.0
116.3
97.4
66.3
55.4
36.5
31.0
21.5
14.4
N9-HEP-HC
rFIX
HC
LC
FXIa activation: - - + +

Figure 7 FIX activity in blood samples from *F9*-KO mice dosed i.v. with 290-360 IU/kg FIX

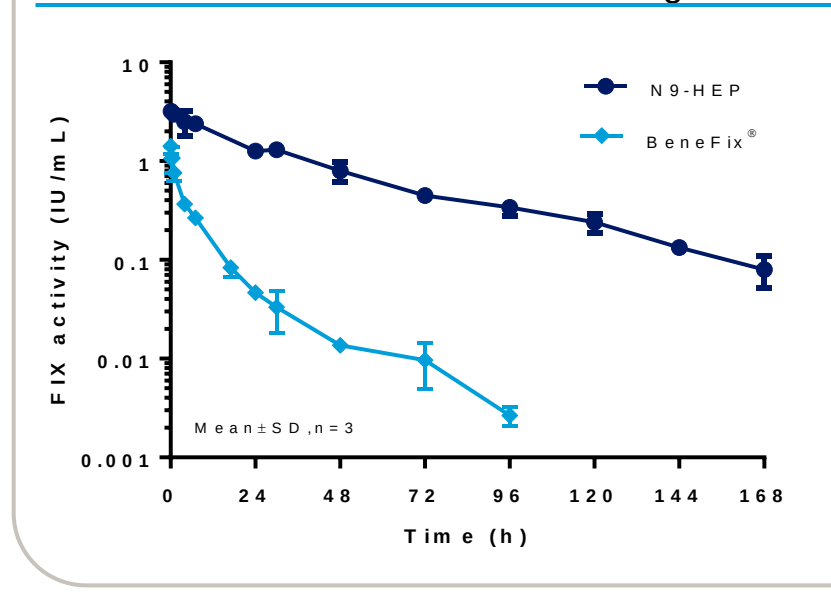
N9-HEP
BeneFix®
FIX activity (IU/mL)
10
1
0.1
0.01
0.001
Mean±SD, n=3
0
24
48
72
96
120
144
168
Time (h)

Table 2 Estimated PK parameters

rFIX	PK parameters		
	t½	CL (mL/h/kg)	MRT (h)
N9-HEP	34	2.5	47
BeneFix®	14	51	12